AXL as a therapeutic target in STK11 mutant NSCLC

#3245

Magnus Blø[1], Austin Rayford[1,2], Noëlly Madeleine[1], Fabian Gärtner[3], Dana Bohan[4], Natalie Ruggio[4], Huiyu Li[5], Luc Girard[5], Rolf Brekken[5], John Minna[5], Marianne Ånerud[3], Wendy Maury[4], Michael Chisamore[6], Claudia Gorcea-Carson[7], Gro Gausdal[1], David Micklem[1], Nigel McCracken[7]

[1]BerGenBio ASA, Bergen Norway, [2]Department of Biomedicine, University of Bergen, Norway, [3]Department of Thoracic Medicine, Haukeland University Hospital, Bergen, Norway
[4]University of Iowa, Iowa City, IA, USA, [5]UT Southwestern Medical Center, Dallas, TX, USA, [6]Merck & Co., Inc., Rahway, NJ, USA, [7]BerGenBio Ltd, Oxford, UK

AACR ANNUAL MEETING 2023, April 14-19, 2023
Orange County Convention Center Orlando, Florida

INTRODUCTION

Serine-threonine kinase 11 (STK11/LKB1) is a tumor suppressor, and loss of STK11 protein contributes to tumorigenesis. Mutations in the STK11 gene (STK11m) are present in ~ 20% of NSCLCs,[1,2] and STK11 is one of the most frequently mutated genes in NSCLC. Inactivation of STK11 is associated with a poorer prognosis, irrespective of treatment modality and represents a sub-group of patients with high unmet need.[3] AXL, a member of the TAM family of receptor tyrosine kinases, is activated in response to inflammation, hypoxia, cellular stress or drug treatment.[4-8] Cancer cells use the AXL pathway to sense stress, triggering molecular mechanisms to ensure survival or escape from the toxic environment (Figure 1). AXL is expressed in both tumor cells, where it enhances survival and drug resistance, and in innate immune cells, such as dendritic cells (DC) and macrophages, where AXL drives immune suppression (Figure 1). The phenotypic characteristics (high energetic and metabolic stress) of a tumor with inactivated STK11 are likely to promote AXL activation in both tumor cells and/or innate immune cells of the tumor microenvironment (TME) (Figure 1). The selective AXL inhibitor bemcentinib targets key survival and resistance mechanisms within the tumor and restores the anti-tumor phenotype of innate immune cells. Specifically, inhibition of AXL in DCs has been shown to increase immune check point inhibition responses in preclinical models of STK11 mutant NSCLC (Figure 2).[22]

Inactivation of STK11 is found in approximately 30% of lung adenocarcinomas as a result of either mutations in the STK11 gene (STK11m) or other non-mutational mechanisms.[2] The Kaufman STK11 loss signature,[2] is a 16 gene classifier for identifying tumors lacking STK11 activity. In the current work, we have applied the Kaufman STK11 loss signature to publicly available data sets (TCGA/CPTAC-3) as well as the BerGenBio trial in 2L NSCLC (BGBC008) to better understand the characteristics and mutational status of these tumors. This signature identifies KEAP1 mutations as strongly associated with STK11 loss. Differential gene expression analysis confirms that mutations in STK11 and KEAP1 have strongly overlapping transcriptional consequences. AXL expression was explored in NSCLC tumors in correlation to STK11m and STK11 loss. Transcriptional changes following AXL inhibition by bemcentinib in a STK11m/KEAP1m NSCLC cell line were mapped to further strengthen the rationale for targeting NSCLC patients characterized by STK11 loss with the AXL inhibitor bemcentinib.

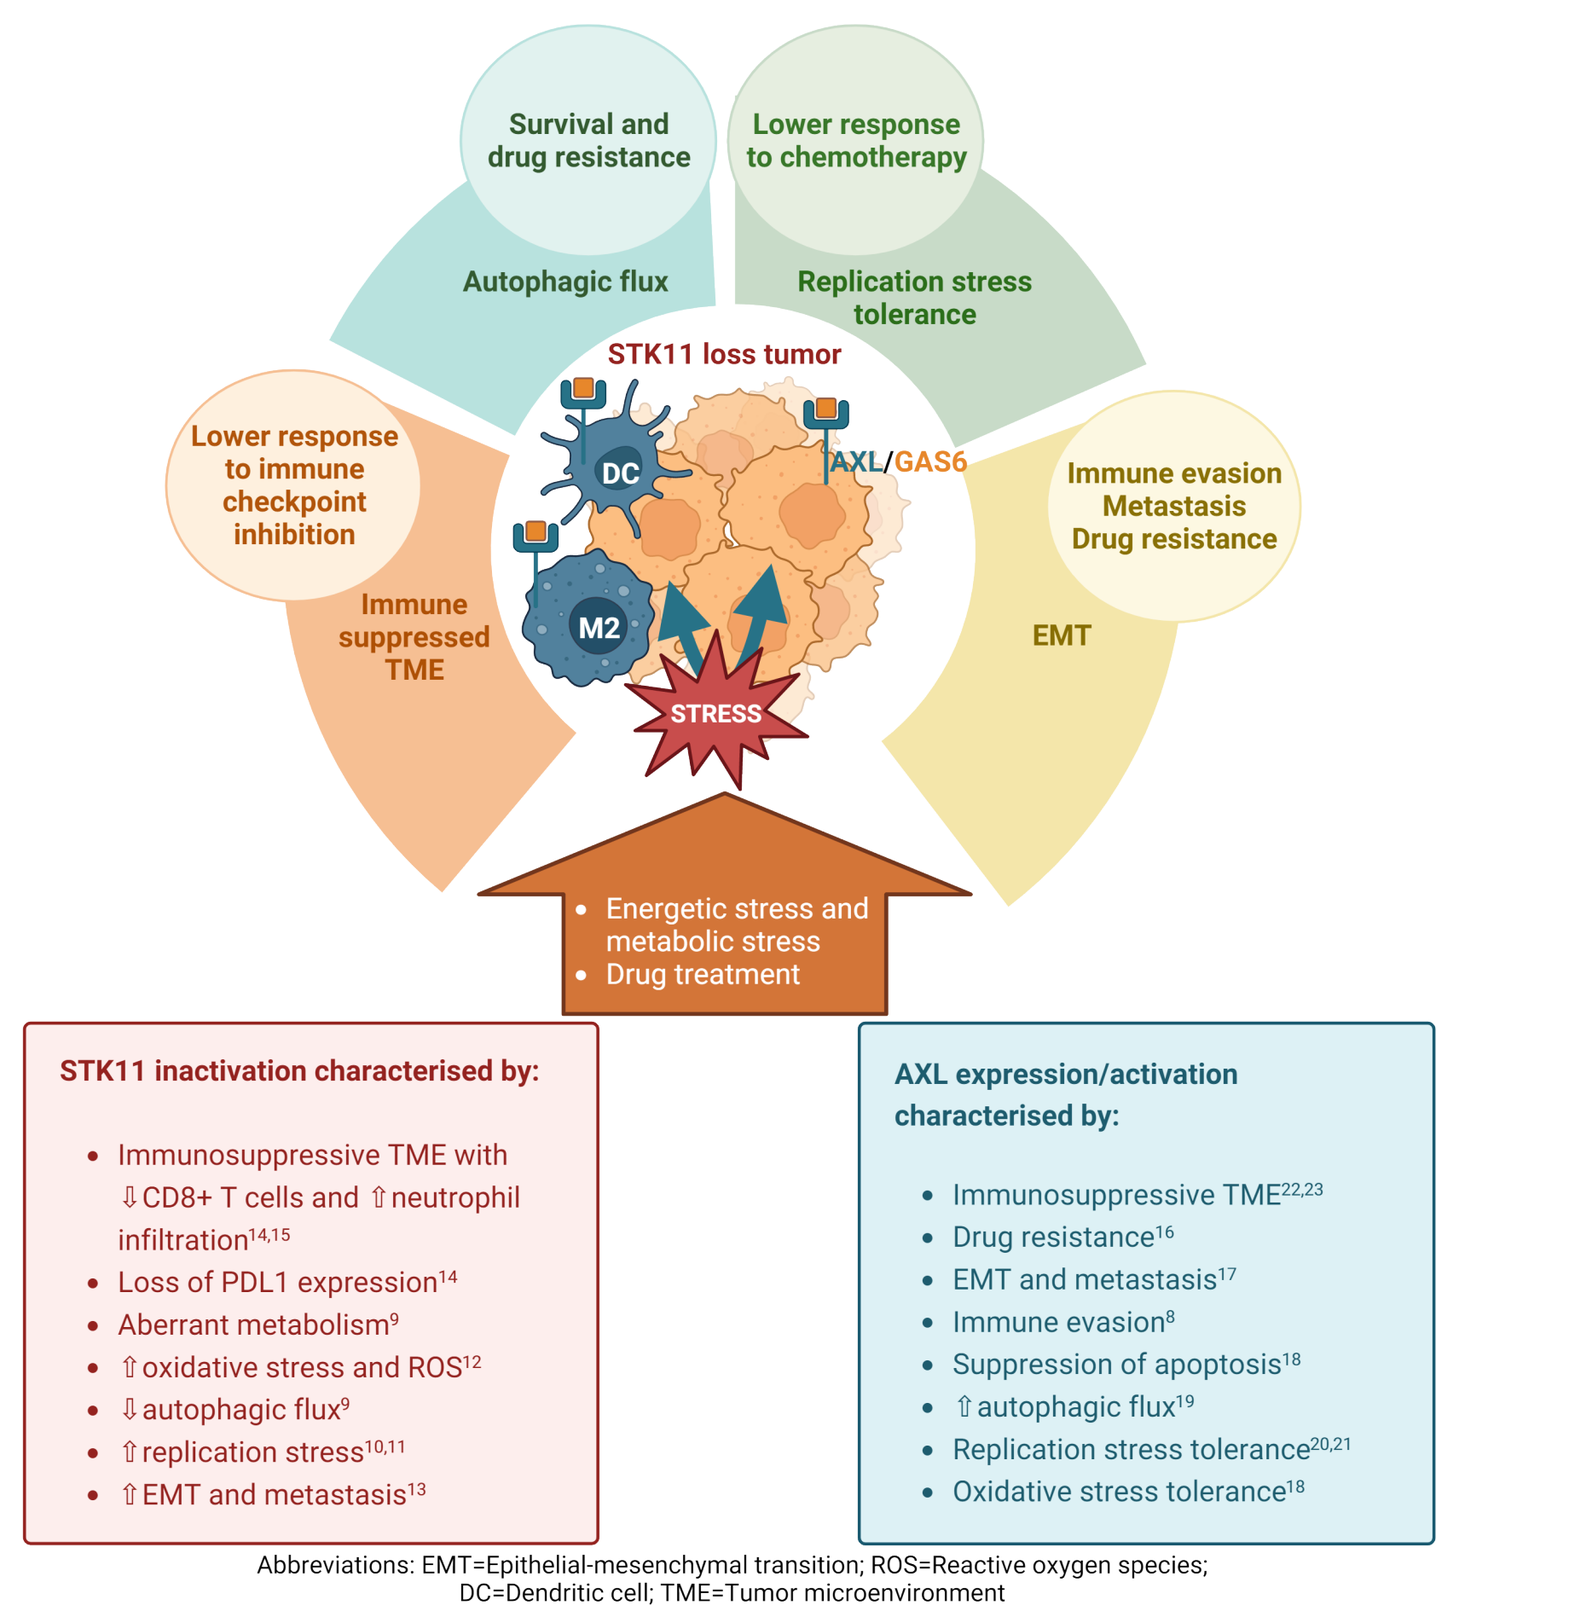
Survival and drug resistance
Lower response to chemotherapy
Autophagic flux
Replication stress tolerance
STK11 loss tumor
Lower response to immune checkpoint inhibition
Immune suppressed TME
DC
M2
AXL/GAS6
Immune evasion Metastasis Drug resistance
EMT
STRESS
Energetic stress and metabolic stress
Drug treatment
STK11 inactivation characterised by:
Immunosuppressive TME with ⇩CD8+ T cells and ⇧neutrophil infiltration[14,15]
Loss of PDL1 expression[14]
Aberrant metabolism[9]
⇧oxidative stress and ROS[12]
⇩autophagic flux[9]
⇧replication stress[10,11]
⇧EMT and metastasis[13]
AXL expression/activation characterised by:
Immunosuppressive TME[22,23]
Drug resistance[16]
EMT and metastasis[17]
Immune evasion[8]
Suppression of apoptosis[18]
⇧autophagic flux[19]
Replication stress tolerance[20,21]
Oxidative stress tolerance[18]

Abbreviations: EMT=Epithelial-mesenchymal transition; ROS=Reactive oxygen species; DC=Dendritic cell; TME=Tumor microenvironment

Figure 1. AXL is a key driver of survival and drug resistance in NSCLC tumors with STK11 loss

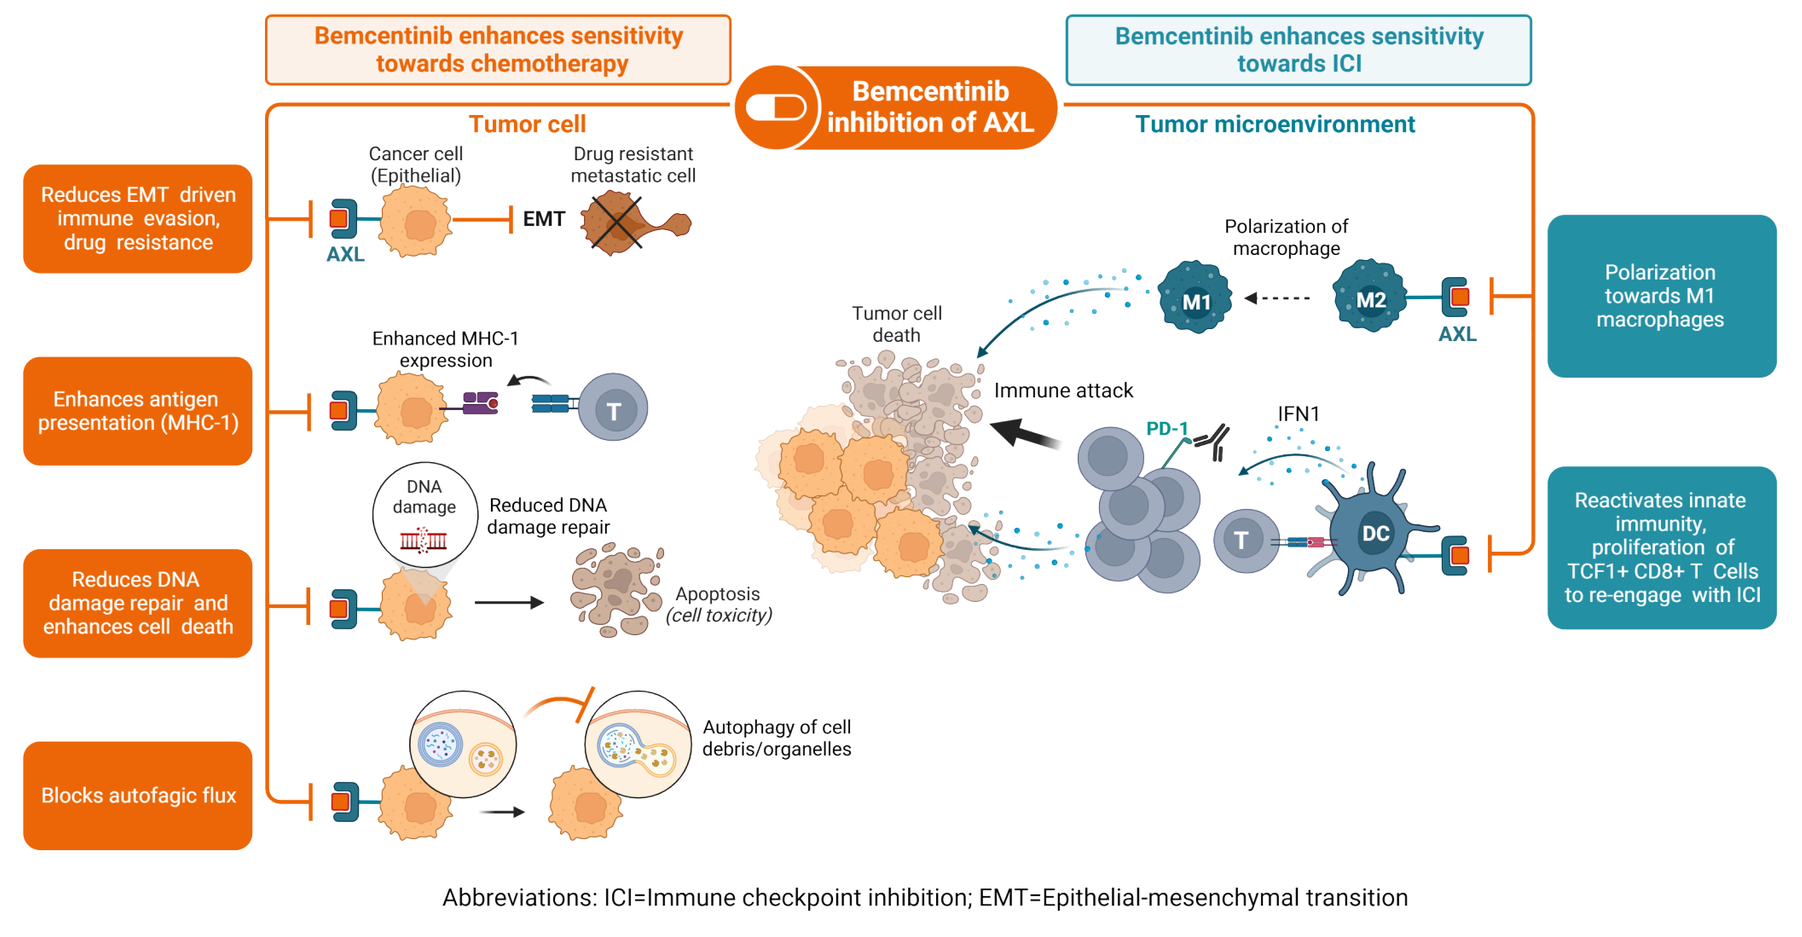
Bemcentinib enhances sensitivity towards chemotherapy
Bemcentinib inhibition of AXL
Bemcentinib enhances sensitivity towards ICI
Tumor cell
Tumor microenvironment
Reduces EMT driven immune evasion, drug resistance
Cancer cell (Epithelial)
Drug resistant metastatic cell
EMT
AXL
Enhances antigen presentation (MHC-1)
Enhanced MHC-1 expression
Reduces DNA damage repair and enhances cell death
DNA damage
Reduced DNA damage repair
Apoptosis (cell toxicity)
Blocks autofagic flux
Autophagy of cell debris/organelles
Polarization of macrophage
M1
M2
Tumor cell death
Immune attack
PD-1
IFN1
T
DC
Polarization towards M1 macrophages
Reactivates innate immunity, proliferation of TCF1+ CD8+ T Cells to re-engage with ICI

Abbreviations: ICI=Immune checkpoint inhibition; EMT=Epithelial-mesenchymal transition

Figure 2. The AXL inhibitor bemcentinib targets key survival and resistance mechanisms of STK11m NSCLC

DATA SOURCES

Clinical Trial NCT03184571 (BGBC008)(†)
- 90 evaluable patients 2L+ NSCLC, 50 with available transcriptomic data.
- Small molecule AXL inhibitor bemcentinib in combination with pembrolizumab.
- RNAseq, whole exome sequencing, AXL IHC, PD-L1 IHC.

Patients from Haukeland University Hospital Bergen REC 45562(‡)[24]
- 119 Stage III/IV NSCLC patients.
- 28% first-line, 72% chemo-refractory 2L+.
- Whole exome sequencing, AXL IHC, PD-L1 IHC.

Public data from Genomic Data Commons (GDC) Data Portal
- 662 lung adenocarcinoma cases with transcriptomic and mutation information.
 - 458 TCGA-LUAD: primary, newly diagnosed, untreated patients.
 - "The results published here are in whole or part based upon data generated by The Cancer Genome Atlas managed by the NCI and NHGRI. Information about TCGA can be found at https://cancergenome.nihgov. dbGaP accession phs000178.v11.p8, retrieved Aug 2022.
 - 204 CPTAC-3: primary, newly diagnosed, untreated patients.
 - "Data used in this publication were generated by the National Cancer Institute Clinical Proteomic Tumor Analysis Consortium. (CPTAC), dbGaP accession phs001287.v16.p6, retrieved Aug 2022.

Patients from Li *et al.*, 2022(*)[22]
- 62 NSCLC cases, known STK11 mutation status, AXL IHC.

METHODS

- AXL IHC staining
 - Staining with 7E10 antibody (Thermo) or C89E7 (Cell Signaling). AXL positive defined as tumor H-score >5 or ≥1% AXL positive immune cells per tumor area.
- PD-L1 IHC staining with 22C3 pharmDx, scored by TPS.
- Whole-exome and mRNA sequencing of FFPE patient samples from BGBC008 and REC 45562 were performed by Almac, Inc.
- Differential gene expression analysis of GDC data using DESeq2, with pathway analysis by Metascape.[26]
- Kaufman STK11 loss score
 - RNAseq count data normalized, variance stabilized and batch-corrected using DESeq2 and limma,[27,28] adjusted by subtraction of mean. Kaufman score calculated as average expression of 16 gene set.[2] Published cut-off at 0.2 captures over 95% of STK11 mutations documented in GDC dataset.
- Circos circle plots: Outer arcs identify each gene list. Inner arcs: dark orange color represents genes that appear in both lists; light orange color represents genes that are unique to that gene list. Purple lines link same genes shared by both gene lists. Blue lines link genes that fall into the same (statistically significant) ontology term.
- Wordclouds generated from GO terms by ranking words by significance of the pathway and frequency.
- GSEA analysis of RNA seq data from A549 cells (STK11m/KEAP1m/KRASm/p5) treated with DMSO or 1 µM bemcentinib for 24 hrs (n=3). Count reads were variance-stabilizing transformed using DESeq2 before GSEA analysis.[29,30]

REFERENCES

1) cBioPortal.org.
2) Kaufman JM., J Thorac Oncol., 2014 Jun;9(6):794-804.
3) Garassino MC., JTO Clin Res Rep., 2022 Nov 8;4(1):100431.
4) Oien DB., Frontiers in Pharmacology, 8, 2018.
5) Quinn JM., Mol Cancer Ther., 2019 Feb;18(2):389-398.
6) Sharif, MN., J Exp Med., 2006 Aug 7; 203(8): 1891–1901.
7) Huang JS., Free Radical Biology and Medicine, 65, 2013, 1246-1256.
8) Terry S., Cancer Immunol Res., 2019 Nov;7(11):1789-1802.
9) Momcilovic M., Br J Cancer, 2015 Aug 11;113(4):574-84.
10) Takahashi N., Cancer Research Communications, 1 June 2022; 2 (6): 503–517.
11) Wang Y., Oncotarget, 8 Nov 20167(45): 73389–73401.
12) Bonanno L., Int J Mol Sci., 2019 Apr 16;20(8):1874.
13) Tzavlaki K., J Cell Physiol., 2023 Feb 15.
14) Skoulidis F., Cancer Discov., 2015;5:860–77.
15) Koyama S., Cancer Res., 2016;76(5):999–1008.
16) Wu F., Int. J. Clin. Exp. Pathol., 2014, 7, 6653–6661.
17) Byers LA., Clin Cancer Res., 2013 Jan 1;19(1):279-90.
18) Liang Z., Apoptosis., 2022 Dec 29.
19) Lotsberg ML., J Thorac Oncol., 2020;S1556-0864(20)30055-1.
20) Balaji K., et al., Molecular cancer research, 2017.
21) Ramkumar K., Mol Cancer Res., 2021 Mar;19(3):485-497.
22) Li H., Cell Rep Med., 2022 Mar 15;3(3).
23) Ludwig KF., Cancer Res., 2018 Jan 1;78(1):246-255.
24) Gärtner F., Acta Oncologica, 2022.
25) Rodríguez-Abreu D., Ann Oncol., 2021 Jul;32(7):881-895.
26) Zhou, Y., Nat Commun., 2019 10, 1523.
27) Love, M.I., Genome Biology, 2014, 15:550.
28) Ritchie ME., Nucleic Acids Research, 2015, 43(7), e47.
29) Subramanian A., Proc Natl Acad Sci., 2005 Oct., 25;102(43):15545-50.
30) Mootha, V., Nat Genet., 2003, 34, 267–273.
31) Hong J., Front Oncol., 2022 Jul 22;12:903874.
32) Krebs M., SITC, 2019.
33) Ramkumar K., AACR 2023 poster #6206.

RESULTS

AXL is expressed in tumor and immune cells in NSCLC patients with or without STK11 loss

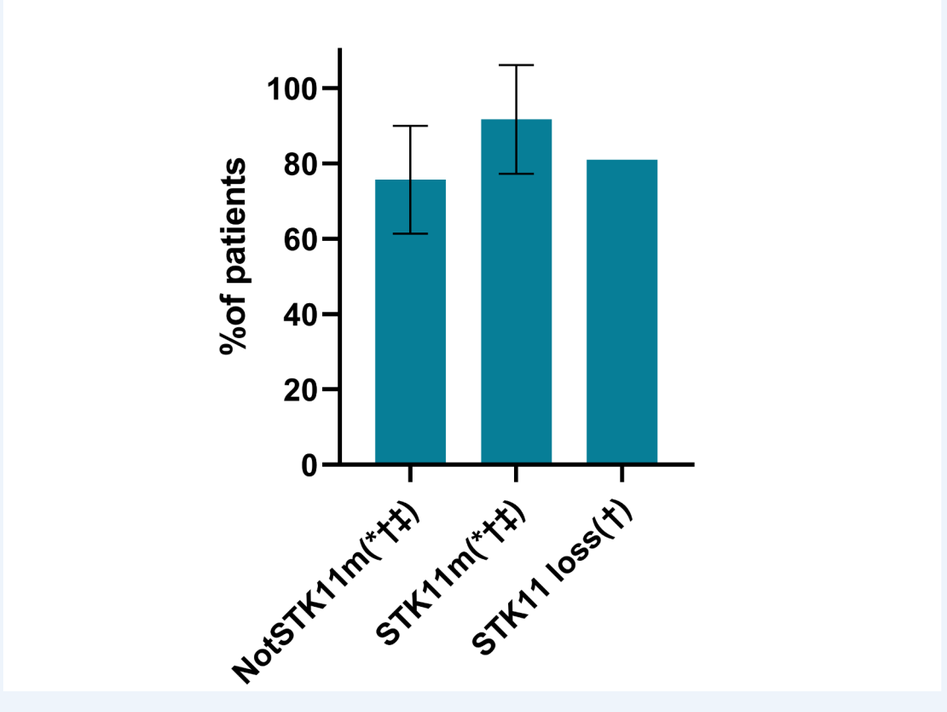
%of patients
100
80
60
40
20
0
NotSTK11m(*†‡)
STK11m(*†‡)
STK11 loss(†)

AXL quantification of IHC from three studies: Li *et al.*, 2022(*),[22] BGBC008(†) and REC 45562(‡).[24] Bars represents AXL positive staining (Tumor H-score >5 or ≥1% immune cells expressing AXL).

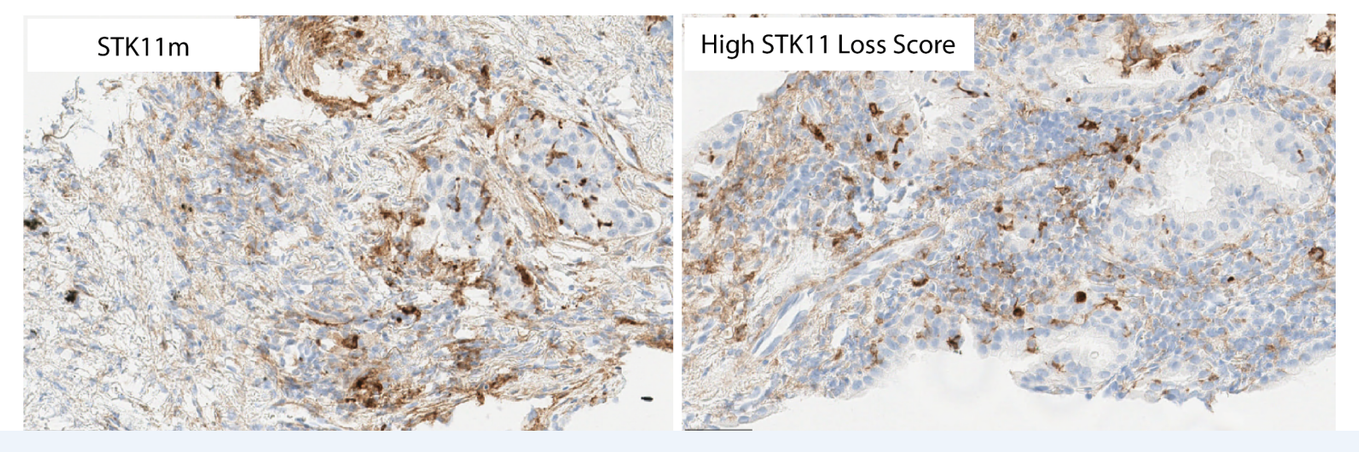
STK11m
High STK11 Loss Score

AXL is expressed on immune and tumor cells in post-chemoimmunotherapy treated NSCLC biopsies(†) with STK11m (left) or high STK11 loss score (right).

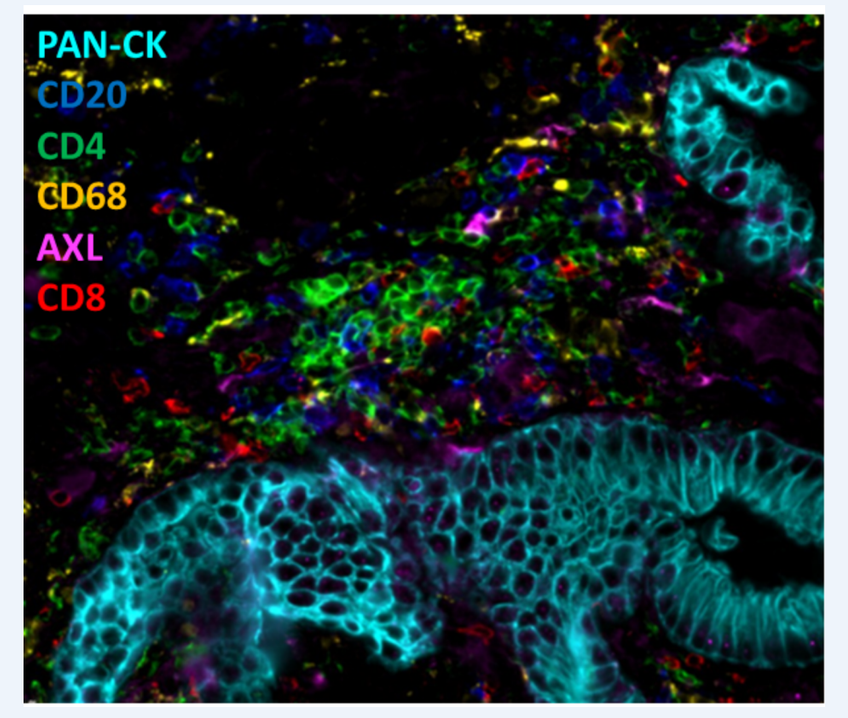
PAN-CK
CD20
CD4
CD68
AXL
CD8

AXL is expressed on tumor associated immune cells and cancer cells in STK11m patient.

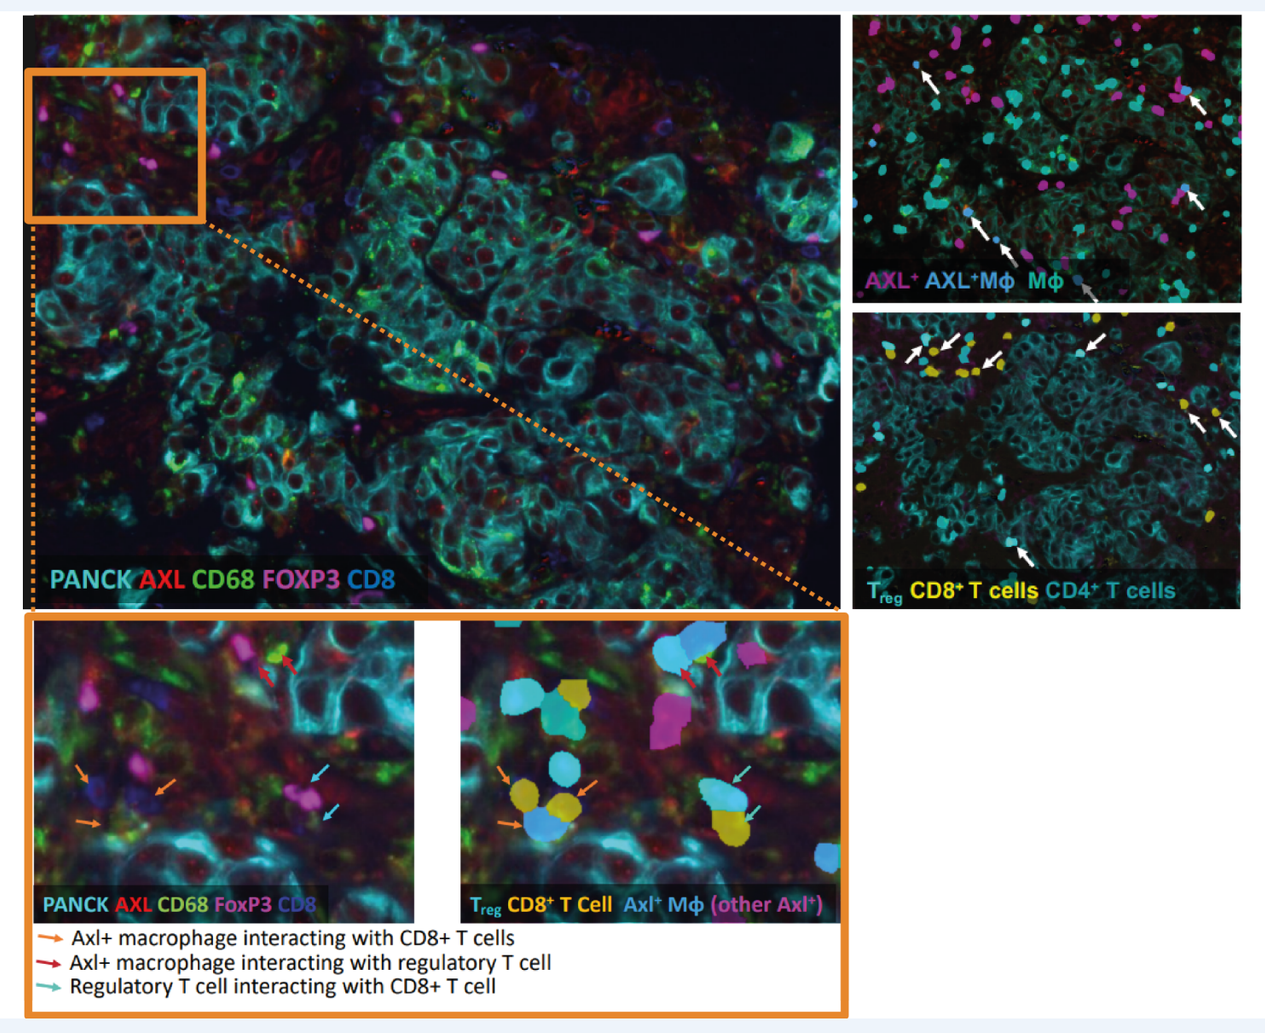
PANCK AXL CD68 FOXP3 CD8
Axl+ macrophage interacting with CD8+ T cells
Axl+ macrophage interacting with regulatory T cell
Regulatory T cell interacting with CD8+ T cell

Tumor infiltrating AXL+ M2 macrophages observed to interact with Tregs and CD8+ T cells in responding patient pretreatment biopsy.[32]

Mutations in STK11 and KEAP1 and low PD-L1 expression are associated with a STK11 loss signature

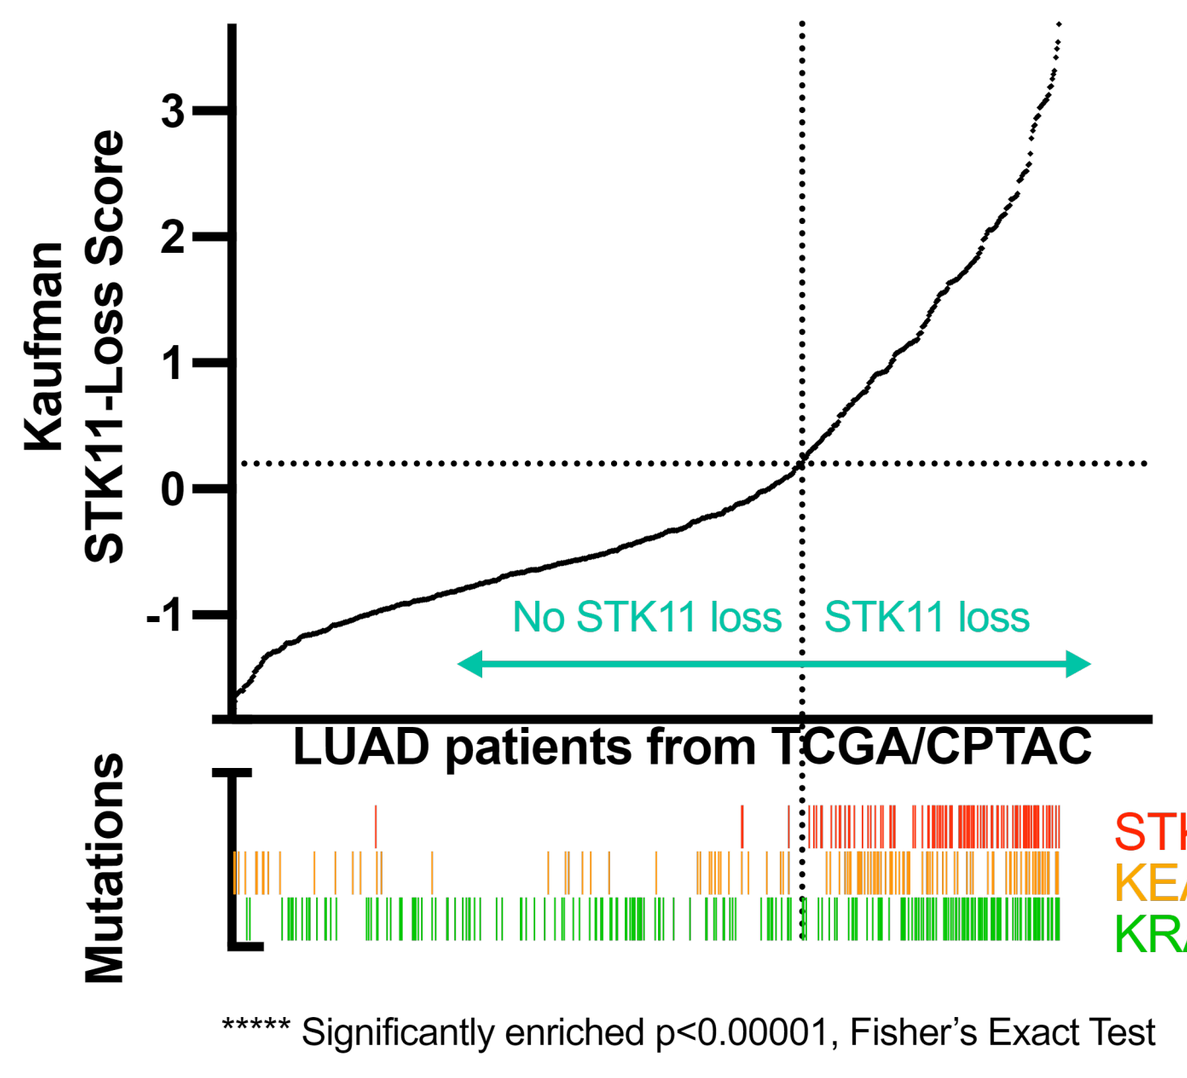
Kaufman STK11-Loss Score
3
2
1
0
-1
No STK11 loss
STK11 loss
LUAD patients from TCGA/CPTAC
Mutations
STK11 *****
KEAP1 *****
KRAS *****
***** Significantly enriched p<0.00001, Fisher's Exact Test

GDC dataset: Patients with the STK11 loss signature are significantly more likely to carry STK11, KEAP1 or KRAS mutations.

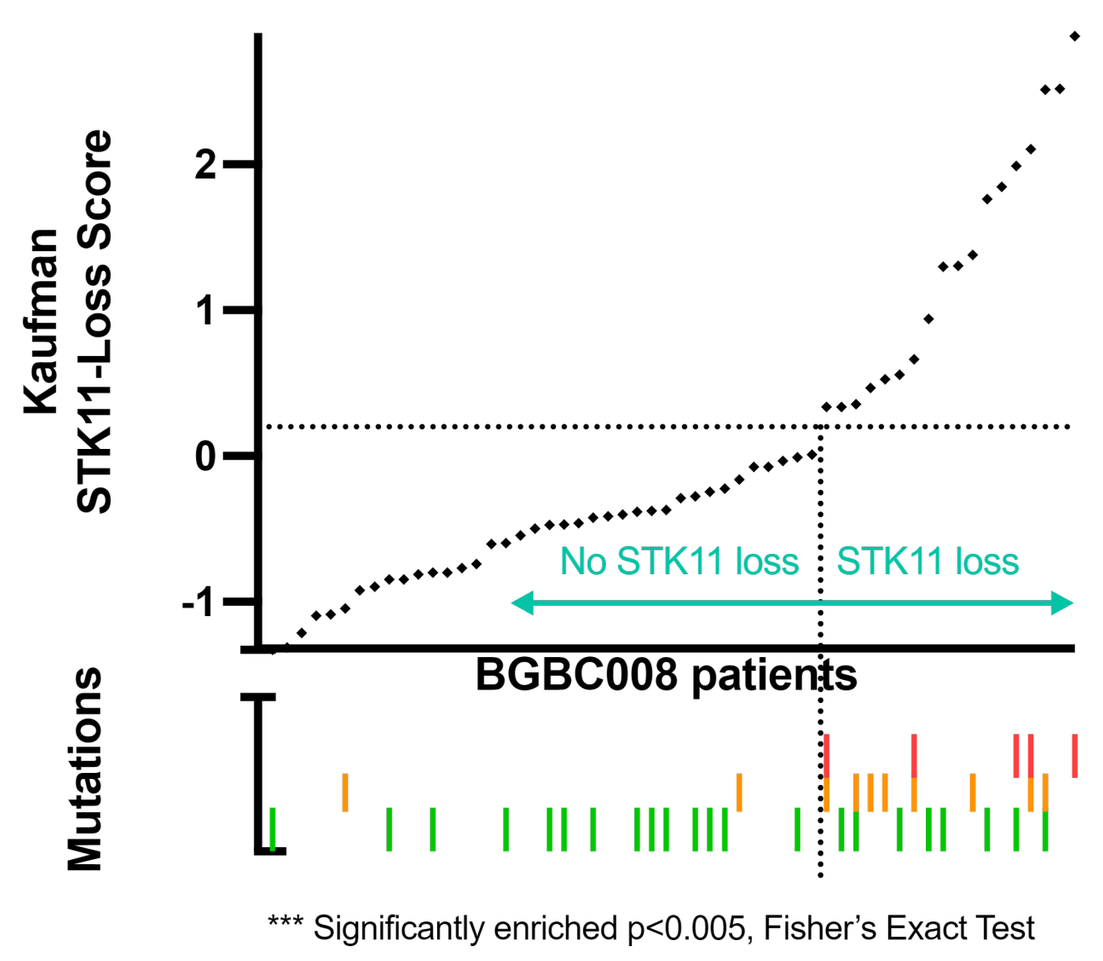
Kaufman STK11-Loss Score
2
1
0
-1
No STK11 loss
STK11 loss
BGBC008 patients
Mutations
STK11 ***
KEAP1 ***
KRAS
*** Significantly enriched p<0.005, Fisher's Exact Test

BGBC008 dataset: Patients with the STK11 loss signature are significantly more likely to carry STK11 or KEAP1 mutations.

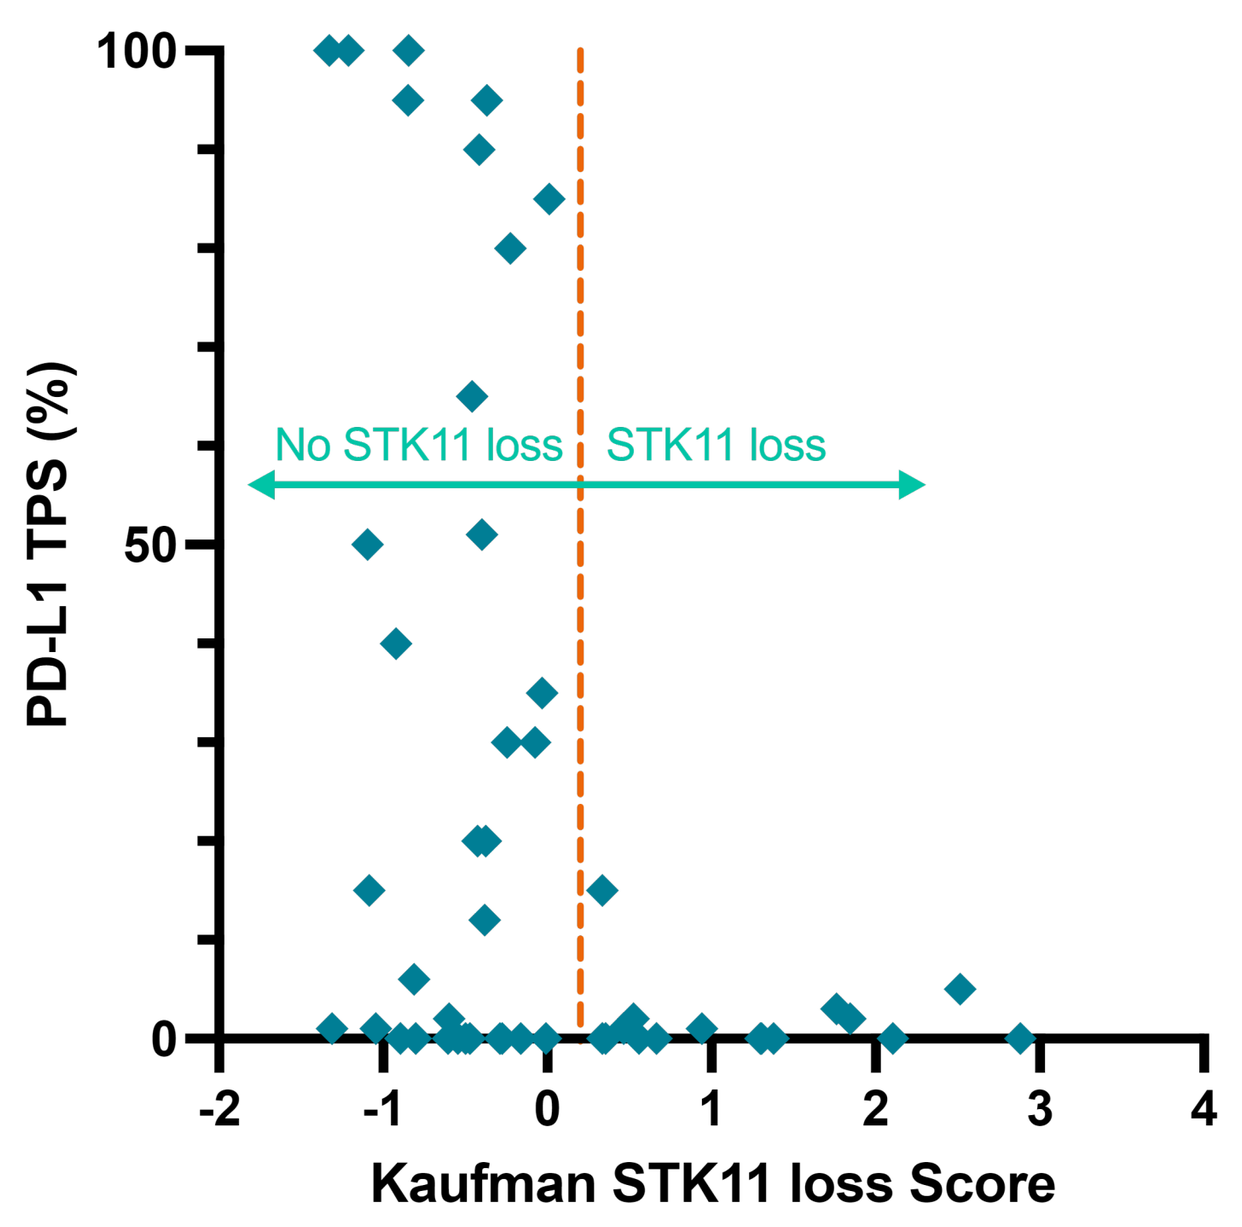
PD-L1 TPS (%)
100
50
0
No STK11 loss
STK11 loss
-2
-1
0
1
2
3
4
Kaufman STK11 loss Score

BGBC008 dataset: Patients with STK11 loss signature are significantly more likely to have low PD-L1 expression (Spearman r=-0.38, p=0.006). These findings are similar to the low PD-L1 expression levels associated with STK11 and KEAP1 mutated patients from the Keynote 189 study.[3]

Mutations in STK11 and KEAP1 promote similar transcriptional consequences in NSCLC patients

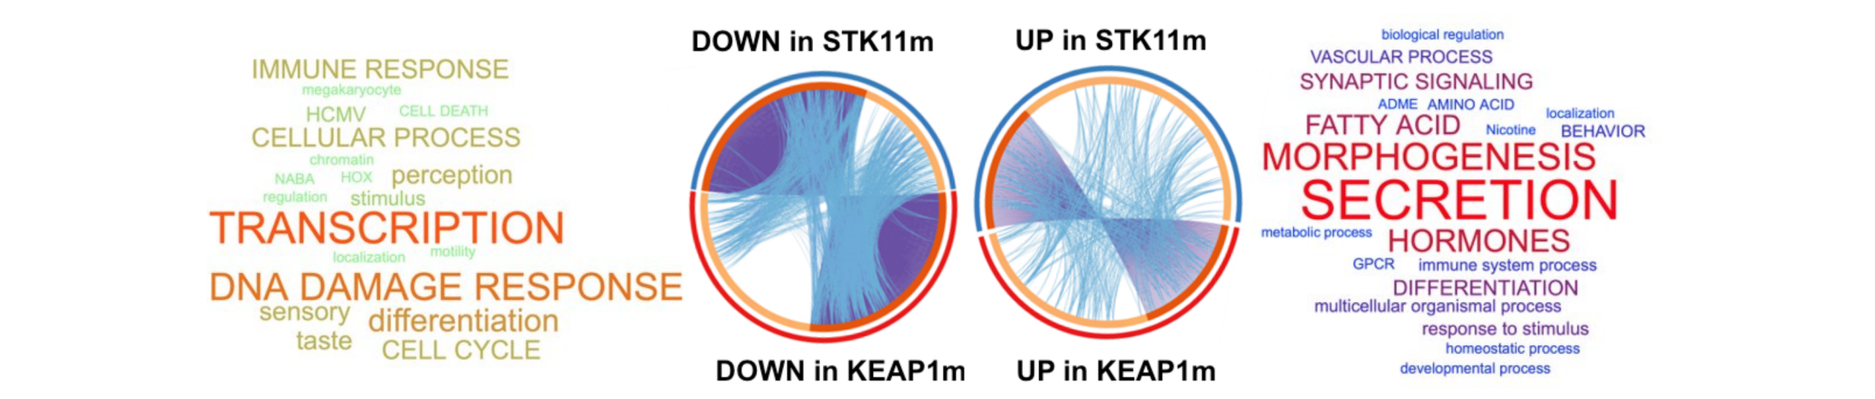
DOWN in STK11m
UP in STK11m
DOWN in KEAP1m
UP in KEAP1m
IMMUNE RESPONSE
CELLULAR PROCESS
perception
TRANSCRIPTION
DNA DAMAGE RESPONSE
sensory
differentiation
taste
CELL CYCLE
VASCULAR PROCESS
SYNAPTIC SIGNALING
FATTY ACID
MORPHOGENESIS
SECRETION
HORMONES
DIFFERENTIATION
multicellular organismal process
response to stimulus
developmental process

- Comparison of differentially-expressed genes from patients carrying STK11m or KEAP1m to patients not carrying the corresponding mutation show a large number of common transcriptional pathways, especially among down-regulated genes.
- Down-regulation of immune response and DNA damage response characteristic of SKT11 tumors.

Bemcentinib modulates pathways involved in tumorigenic features of STK11m NSCLC

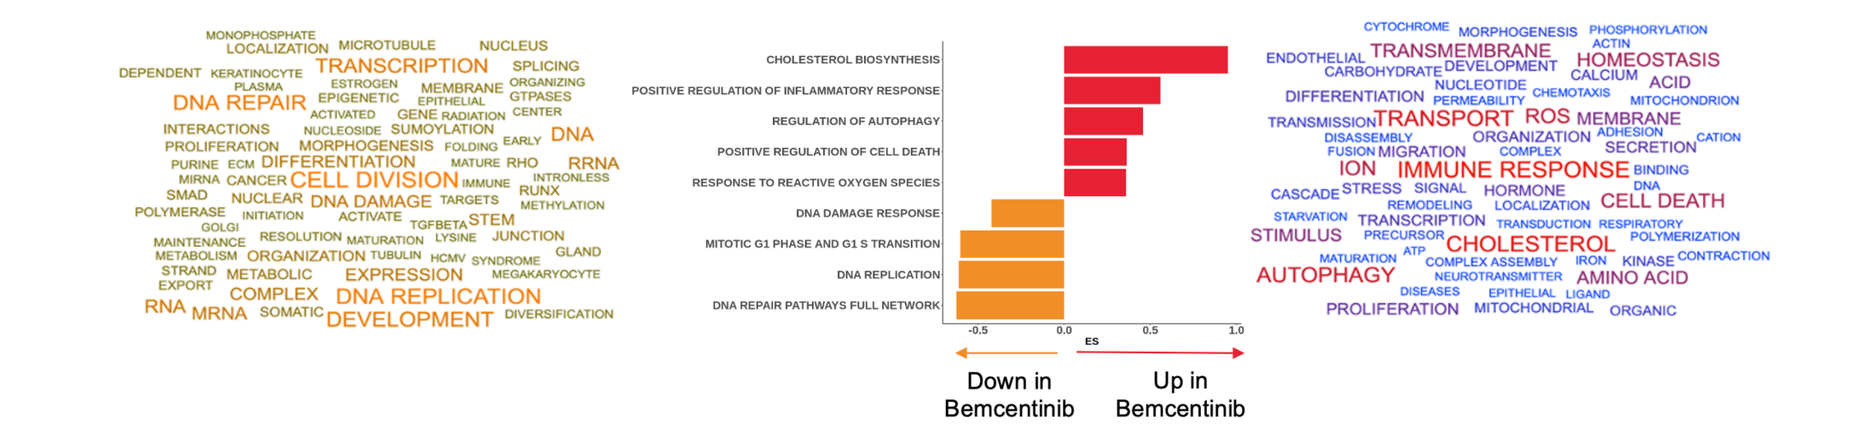
CHOLESTEROL BIOSYNTHESIS
POSITIVE REGULATION OF INFLAMMATORY RESPONSE
REGULATION OF AUTOPHAGY
POSITIVE REGULATION OF CELL DEATH
RESPONSE TO REACTIVE OXYGEN SPECIES
DNA DAMAGE RESPONSE
MITOTIC G1 PHASE AND G1 S TRANSITION
DNA REPLICATION
DNA REPAIR PATHWAYS FULL NETWORK
Down in Bemcentinib
Up in Bemcentinib
TRANSCRIPTION
DNA REPAIR
CELL DIVISION
DNA DAMAGE
DNA REPLICATION
TRANSMEMBRANE
HOMEOSTASIS
TRANSPORT
IMMUNE RESPONSE
CELL DEATH
CHOLESTEROL
AUTOPHAGY

Inhibition of AXL in A549 NSCLC cells (STK11m/KEAP1m) with bemcentinib upregulates inflammatory response signatures, modulates autophagy, and down-regulates pathways required for survival in response to replication stress and DNA damage (see also AACR Poster 6206).[33]
A549 cells (STK11m/KEAP1m/KRASm/p53wt) treated with bemcentinib or DMSO. Center: Selected enriched terms (GSEA). Left/Right: Word clouds of enriched terms (Right, blue/red: enriched in bemcentinib treated; Left, green/orange: enriched in control).

Bemcentinib in combination with pembrolizumab in 2L+ NSCLC has similar activity when STK11 is present and absent

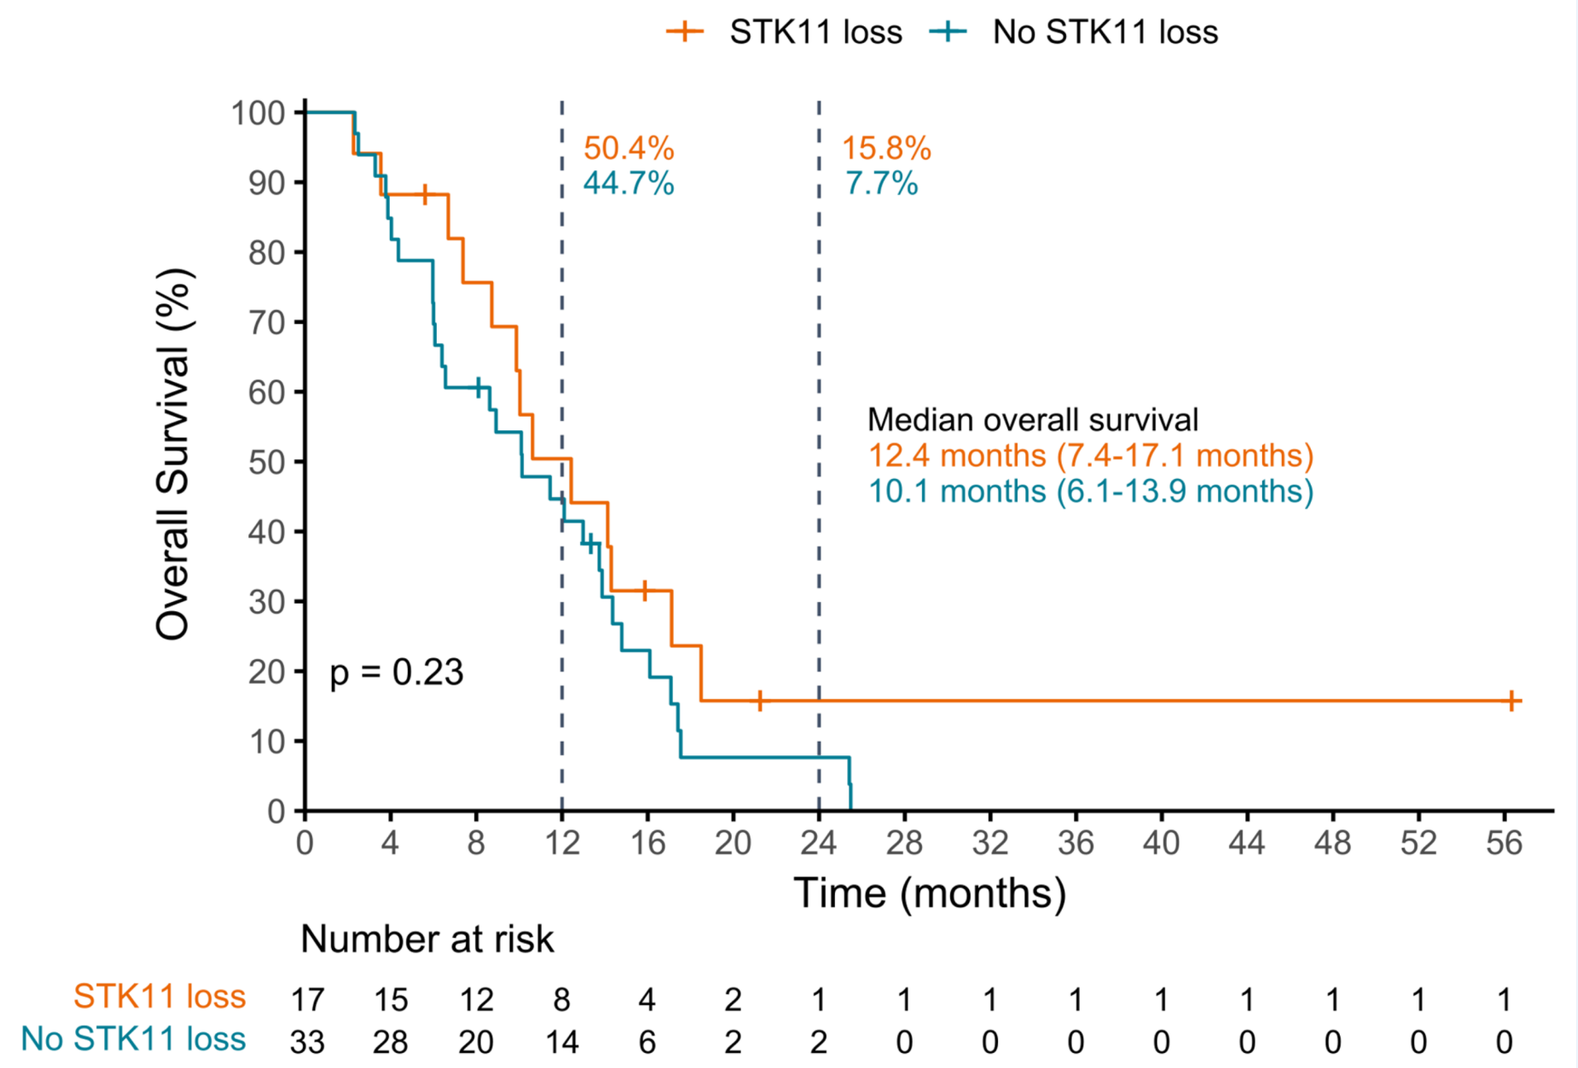
STK11 loss
No STK11 loss
Overall Survival (%)
50.4%
44.7%
15.8%
7.7%
Median overall survival
12.4 months (7.4-17.1 months)
10.1 months (6.1-13.9 months)
p = 0.23
Time (months)
Number at risk
STK11 loss 17 15 12 8 4 2 1 1 1 1 1 1 1 1 1
No STK11 loss 33 28 20 14 6 2 2 0 0 0 0 0 0 0 0

Comparison of Overall Survival BGBC008
No significant difference between STK11 loss and no STK11 loss.

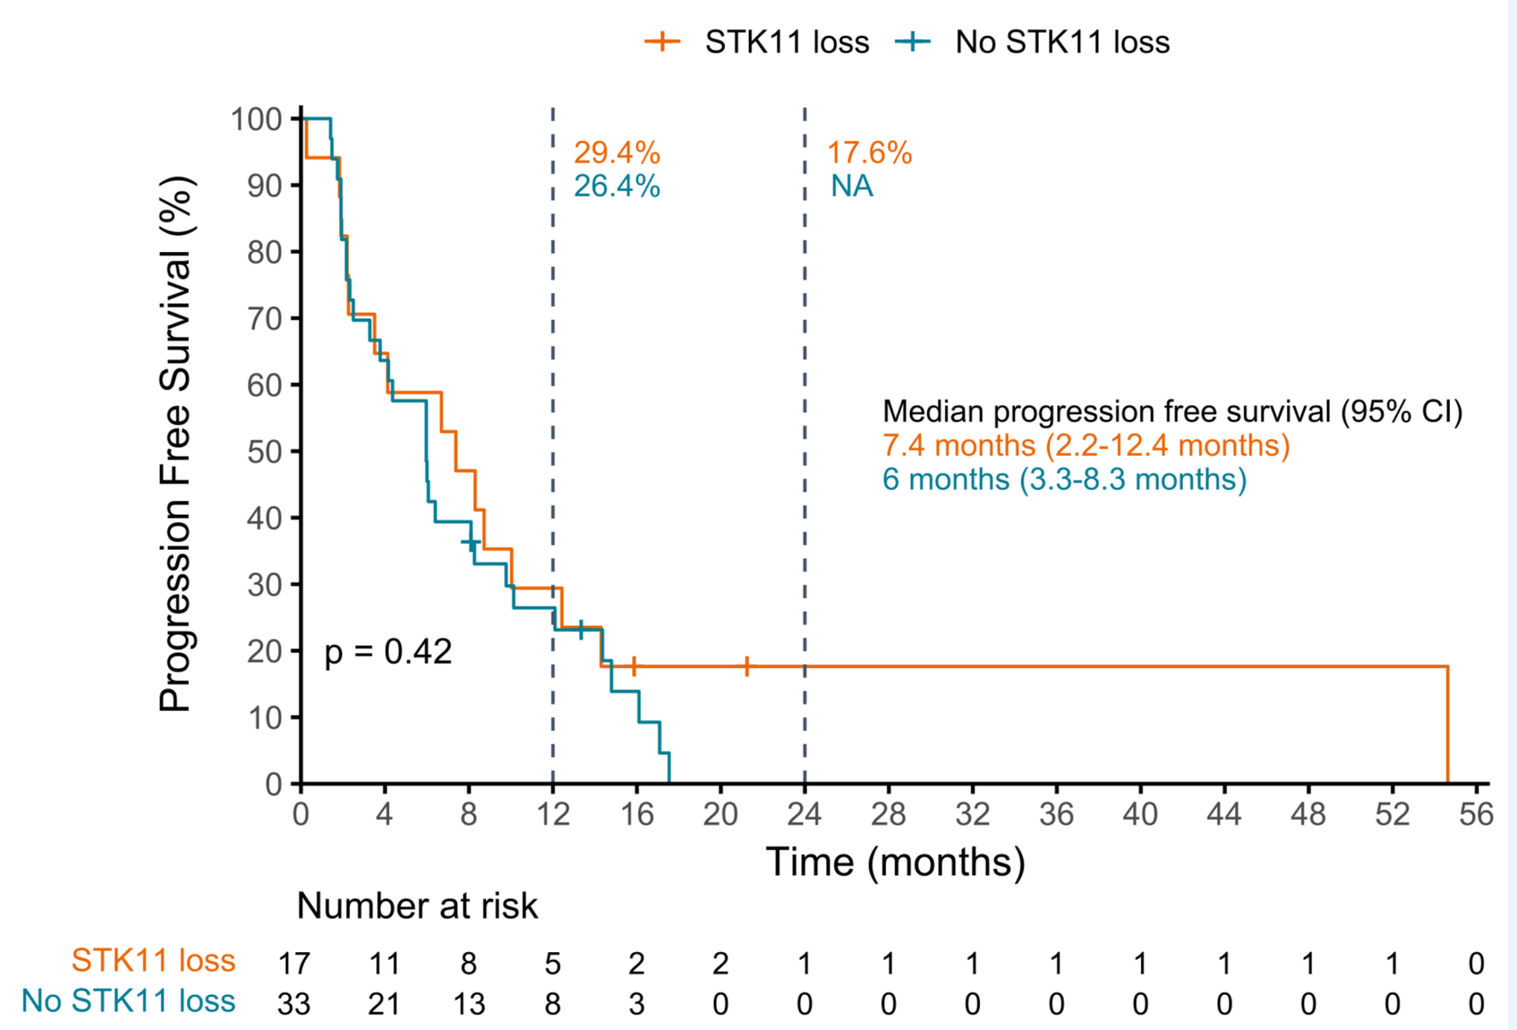
STK11 loss
No STK11 loss
Progression Free Survival (%)
29.4%
26.4%
17.6%
NA
Median progression free survival (95% CI)
7.4 months (2.2-12.4 months)
6 months (3.3-8.3 months)
p = 0.42
Time (months)
Number at risk
STK11 loss 17 11 8 5 2 2 1 1 1 1 1 1 1 1 0
No STK11 loss 33 21 13 8 3 0 0 0 0 0 0 0 0 0 0

Comparison of Progression Free Survival BGBC008
No significant difference between STK11 loss and no STK11 loss.

Bemcentinib + pembrolizumab combination compares favorably to existing therapies in 2L NSCLC

	pemetrexed-platinum		Pembrolizumab + pemetrexed-platinum	
First Line[3]	OS	PFS	OS	PFS
Keynote 189 STK11 WT	12.4 (7.6-25.0)	4.7 (4.6-5.3)	22.5 (20.1-NR)	10.4 (8.2-14.0)
Keynote 189 KEAP1 WT	12.2 (7.6-25.0)	4.7 (4.5-5.3)	23.5 (20.4-NR)	10.4 (8.2-14.0)
Keynote 189 STK11 mutated	8.4 (6.5-NR)	4.8 (4.5-8.6)	17.4 (4.7-NR)	6.1 (3.6-9.2)
Keynote 189 KEAP1 mutated	8.7 (6.6-NR)	4.8 (4.6-8.8)	13.3 (7.5-NR)	5.1 (3.7-11.1)

STK11/KEAP1 mutations reduce survival irrespective of treatment

Pembrolizumab provides survival benefit to all groups

Second Line +	Pembrolizumab monotherapy	
	OS	PFS
Keynote 189 control crossover[25]*	6.9 (4.8-10.5)	2.8 (2.5-2.9)
	Bemcentinib + Pembrolizumab	
BGBC008 No STK11 loss	10.1 (6.1-13.9)	6.0 (3.3-8.3)
BGBC008 STK11 loss	12.4 (7.4-17.1)	7.4 (2.2-12.4)

Bemcentinib provides similar clinical outcomes

*On study crossover of placebo plus pemetrexed-platinum group (n=84) to pembrolizumab monotherapy

Clinical outcomes from the Phase 2, 2L+ metastatic NSCLC study (BGBC008) investigating the combination of bemcentinib and pembrolizumab indicate that patients with STK11 loss have similar clinical outcomes to non-STK11 loss patients.

CONCLUSIONS

- STK11/KEAP1 mutations are transcriptionally similar and share a common signature for STK11 loss.
- AXL is a recognized driver of key features characterizing STK11 inactivated tumors such as an immune suppressed TME and tumor drug resistance and metastasis.
- AXL is expressed in over 80% of NSCLC tumors that show STK11 loss/mutation.
- STK11 loss due to mutations in the STK11 gene (STK11m) or other mechanisms such as KEAP1m is associated with low level of PD-L1 expression and poorer outcomes.
- Clinical outcomes from the Phase 2, 2L+ metastatic NSCLC study (BGBC008) investigating the combination of bemcentinib and pembrolizumab indicate that patients with STK11 loss have similar clinical outcomes to non-STK11 loss patients, providing a rationale to target NSCLC with mutations in either STK11 or KEAP1 with an AXL inhibitor.
- The clinical utility of targeting AXL in patients with STK11 mutation will be further explored in a global, open-label Phase 1b/2a trial of bemcentinib with SOC (pembrolizumab, pemetrexed and carboplatin) in 1L advanced/metastatic non-squamous NSCLC patients [NCT05469178].

ACKNOWLEDGEMENTS

This study is in collaboration with Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA.
Thanks to all patients and their families, investigators and study coordinators who participated in and supported this study.

CONTACT INFORMATION

BerGenBio ASA
Møllendalsbakken 9
5009 Bergen, Norway

post@bergenbio.com
www.bergenbio.com
Twitter: @BGenBio

Created with BioRender Poster Builder